European Treatment and Outcome Study (EUTOS) for Chronic Myeloid Leukemia (CML): A Public/Private Partnership of Academia and Industry

Kossak, U.[1], Rohrbacher, M.[1]; Baccarani, M.[2], Guilhot, J.[3], Hasford, J.[4], Simonsson, B.[5], Engwall, AL.[6], P. Schuld[6]; Cross, NCP.[7], Hochhaus, A.[1], Saglio, G.[8], A. Covelli[6]; Mahon, FX.[9], F. Guilhot[3]; Cervantes, F.[10], Saussele, S.[1], Pane, F.[11], L. Montrucchio, L.[6], R. Hehlmann[1]

[1] Medizinische Fakultät Mannheim, Universität Heidelberg, Germany, [2] Università di Bologna, Italy, [3] Université de Poitiers, France, [4] LM Universität München, Germany, [5] University Hospital Uppsala, Sweden, [6] Novartis Pharma, Basel, Switzerland, [7] University of Southampton, UK, [8] Università di Torino, Italy, [9] Université de Bordeaux, France, [10] Hospital Clinic Provincial de Barcelona, Spain, Università di Napoli, Italy

Introduction

The **European Treatment and Outcome Study (EUTOS) for Chronic Myeloid Leukemia (CML) –** a unique collaboration between industry (Novartis) and academia (European LeukemiaNet, ELN) – has been launched in summer 2007. The collaboration will run for 3 years, from June 2007 until spring 2010, with a possibility to extend the contract between Novartis and the University of Heidelberg (legal representative of the ELN-CML group).

Objectives

EUTOS aims to:

- Enhance the understanding of the nature and management of CML
- Improve standardized evaluation and monitoring and provide quality-controlled outcomes for CML
- Optimize diagnosis and treatment of CML across Europe

Four subprojects support these aims:

- Expanded CML Registry
- Molecular Monitoring
- Pharmacological Monitoring
- Spread of Excellence

Figure 1: Kick-off Meeting in June 2007

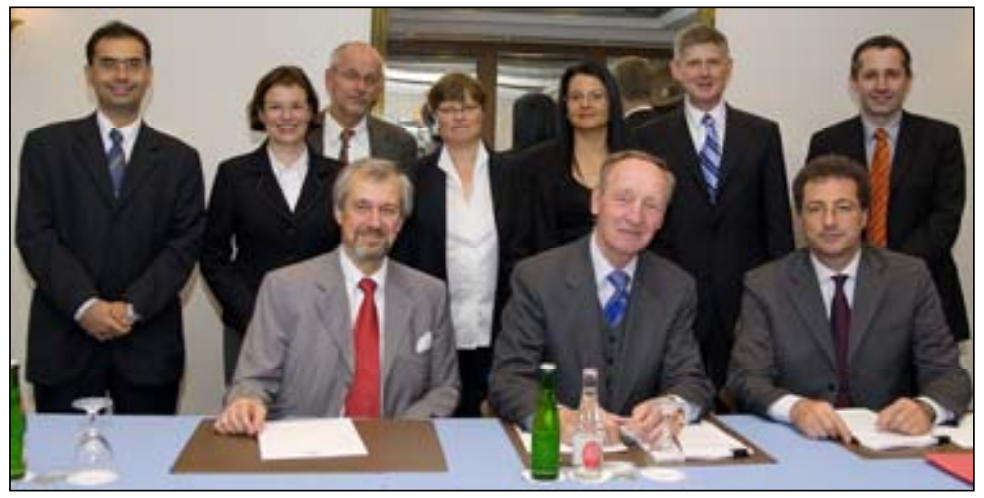

Goals of the subprojects

Expanded CML Registry

Collection of baseline, treatment and outcome data:

- Determination of variations according to demographic and geographical differences
- Evaluation of quality-controlled outcomes and implementation of ELN recommendations
- Development of a comprehensive prognostic model to optimize leukemia treatment

Molecular Monitoring

RQ-PCR standardization:

- Distribution of standardized RQ-PCR technology for BCR-ABL quantification and mutation detection across Europe
- Determination of the prognostic significance of mutations
- Establishment of ELN-certified labs that can serve as a national reference centres

Pharmacological Monitoring

Imatinib blood level testing:

- Standardization of imatinib blood level testing throughout Europe
- Expansion of the availability of free of charge imatinib BLT for European patient blood samples
- Establish monitoring facilities in respective countries, using a standardized monitoring protocol
- Construction of a reliable dosing database

Spread of Excellence

Education and PR:

- Bring together all EUTOS for CML subprojects into a clearly defined offering for physicians
- Ensure high-quality medical education and resources for all physicians caring for patients with CML
- Monitoring of the implementation of the official ELN guidelines

Achievements

To reduce bias, the Registry is divided into three patient groups:

- In-study: 1900 patients enrolled in prospective studies, imatinib frontline
- Out-study: 1560 patients registered in databases, irrespective of frontline treatment
- Prospective: 1700 newly diagnosed patients not previously in registries or clinical studies (from 2009)

International standardization process in 50 centres is ongoing:

- More than a half of these centres have already received samples
- National reference laboratories are being set up
- Educational documents describing project and providing lab guidelines have been circulated

National reference centres are being set up in France, Italy, Germany, and Spain:

- A laboratory kit has been created to facilitate sample submission and to ensure that all samples arrive in quality and time
- Free shipping and testing of patient blood samples for imatinib plasma-level determination is available for physicians in Europe

Materials to support the implementation of the EUTOS for CML project:

- Physician information pack to support the Pharmacological Monitoring project
- Dedicated ELN booth for display at major congresses
- ELN recommendations summary card
- Website, Flyer, Posters, Slide kit, Newsletter

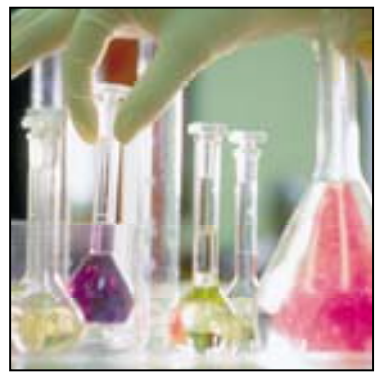

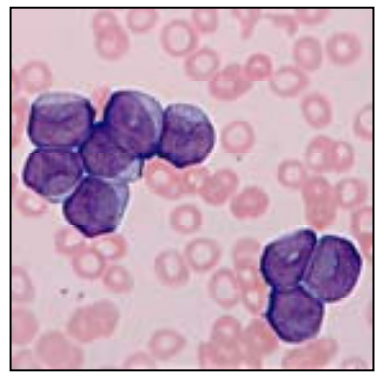

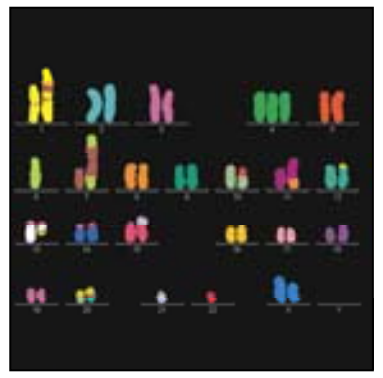

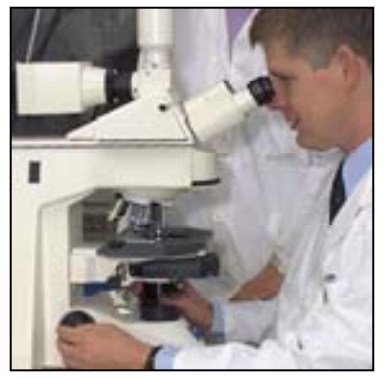

Figure 2: Molecular Monitoring. Interlaboratory results before and after conversion

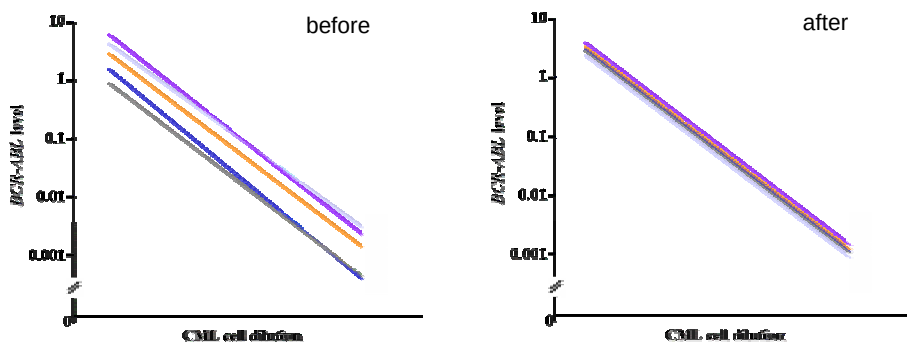
before
after
BCR-ABL level
10
1
0.1
0.01
0.001
0
CML cell dilution

Differences in testing methods and control genes have caused variability between laboratories, making it difficult to compare results for the same patient sample measured in different laboratories. Standardization of RQ-PCR methodology and reporting allows the results from different laboratories to be compared with one another.

Figure 3: Pharmacological Monitoring. Patients not responding as well as expected

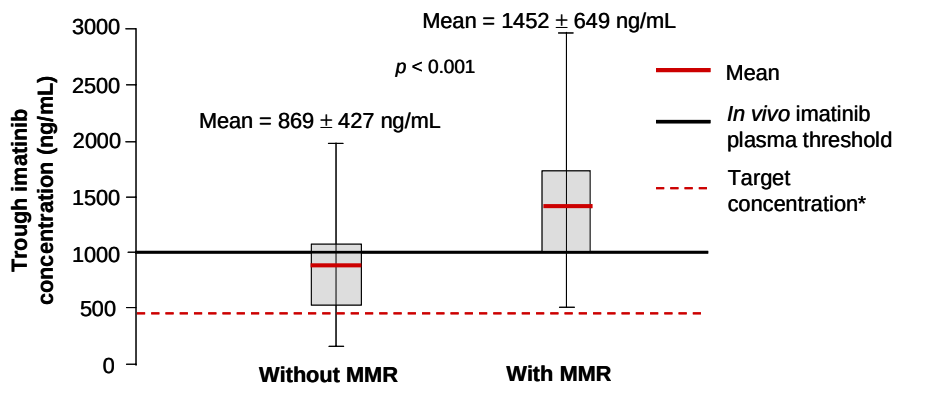
Trough imatinib concentration (ng/mL)
3000
2500
2000
1500
1000
500
0
Mean = 869 ± 427 ng/mL
Mean = 1452 ± 649 ng/mL
p < 0.001
Without MMR
With MMR
Mean
In vivo imatinib plasma threshold
Target concentration*

*Required to result in *BCR-ABL*–positive cell death *in vitro*.

Picard *et al. Blood* 2007;109:3496–3499.

Mean trough imatinib level was significantly higher in patients who achieved MMR than in those who did not. The study identified a plasma ‘threshold’ of 1002 ng/mL: the majority (76%) of patients with MMR had trough plasma levels exceeding the 1002 ng/mL threshold, while 71% of patients without MMR had trough plasma levels below the threshold.

Financial Disclosure: No conflicts.
EUTOS is funded by the EU and an unrestricted grant from Novartis.